Interdisciplinary Approach to Human-Centered COVID-19 Communication — Learning from Explainable AI

Jasminko Novak[1,2], Tina Maljur[1], Mathias Becker[1], Maja Novak[1]

[1] European Institute for Participatory Media, Berlin, Germany

[2] Stralsund University of Applied Sciences, Stralsund, Germany

Funded by: VolkswagenStiftung

Background & Motivation

Crises such as the COVID-19 pandemic present many complex challenges, one of which is an "**explanation problem**" in communicating scientific findings and complex societal decisions. The general public is confronted with expert recommendations and political decisions that are the result of **more or less transparent**, **complex** processes, the rationales of which are often not readily apparent and understandable to non-experts, which is all happening in a period of high uncertainty. This undermines the **trustworthiness** and **acceptance** of these decisions. Persistent, widespread misinformation further complicates communication. This creates a disconnect between expert communication (even when it is clear, transparent, and evidence-based) and the public's more intuitive reasoning.

Goal

To **design a solution** that aims to **innovatively solve the "explanation problem"**, by adapting an **interdisciplinary**, **human-centered approach**, building on findings from the **explainable AI** (also dealing with a complex, in-transparent system that is hard to fathom for non-experts), learning from **psychology**, **communication studies** and **human-computer interaction**.

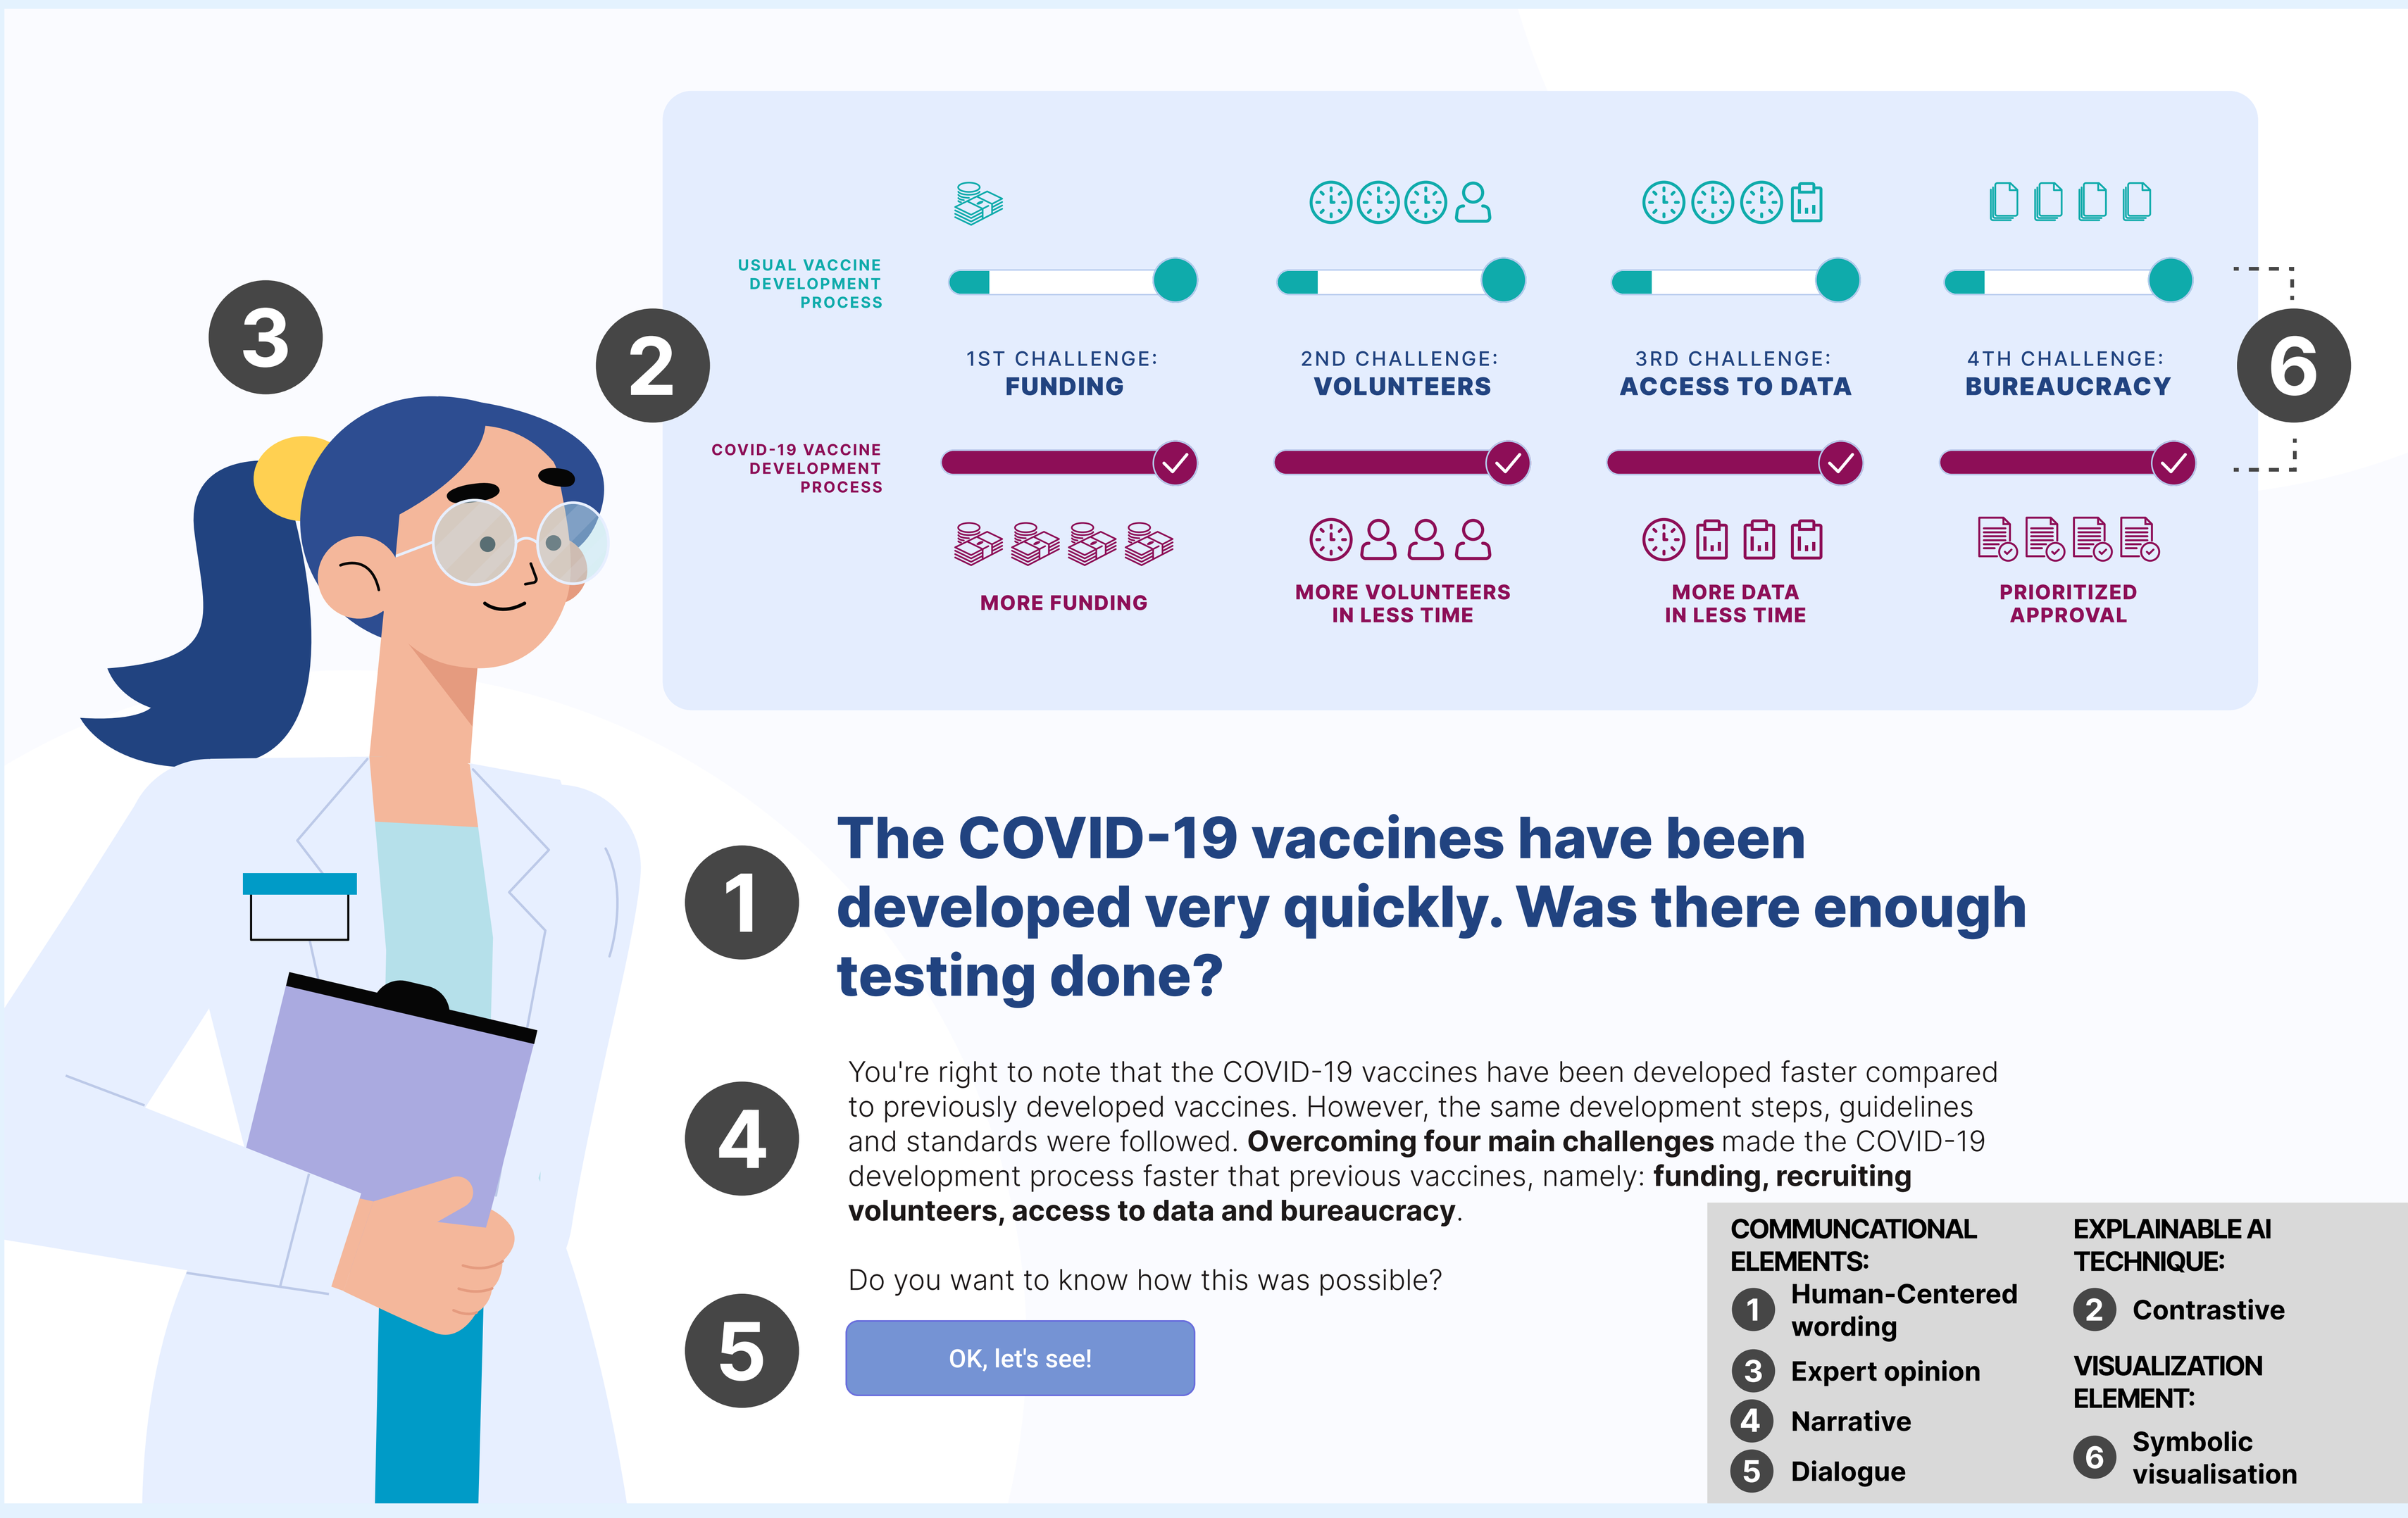

USUAL VACCINE DEVELOPMENT PROCESS
1ST CHALLENGE: FUNDING
2ND CHALLENGE: VOLUNTEERS
3RD CHALLENGE: ACCESS TO DATA
4TH CHALLENGE: BUREAUCRACY
COVID-19 VACCINE DEVELOPMENT PROCESS
MORE FUNDING
MORE VOLUNTEERS IN LESS TIME
MORE DATA IN LESS TIME
PRIORITIZED APPROVAL
The COVID-19 vaccines have been developed very quickly. Was there enough testing done?
You're right to note that the COVID-19 vaccines have been developed faster compared to previously developed vaccines. However, the same development steps, guidelines and standards were followed. Overcoming four main challenges made the COVID-19 development process faster that previous vaccines, namely: funding, recruiting volunteers, access to data and bureaucracy.
Do you want to know how this was possible?
OK, let's see!
COMMUNCATIONAL ELEMENTS:
1 Human-Centered wording
3 Expert opinion
4 Narrative
5 Dialogue
EXPLAINABLE AI TECHNIQUE:
2 Contrastive
VISUALIZATION ELEMENT:
6 Symbolic visualisation

Figure 1. The initial screen of the prototype with elements of the explanation mapped out.

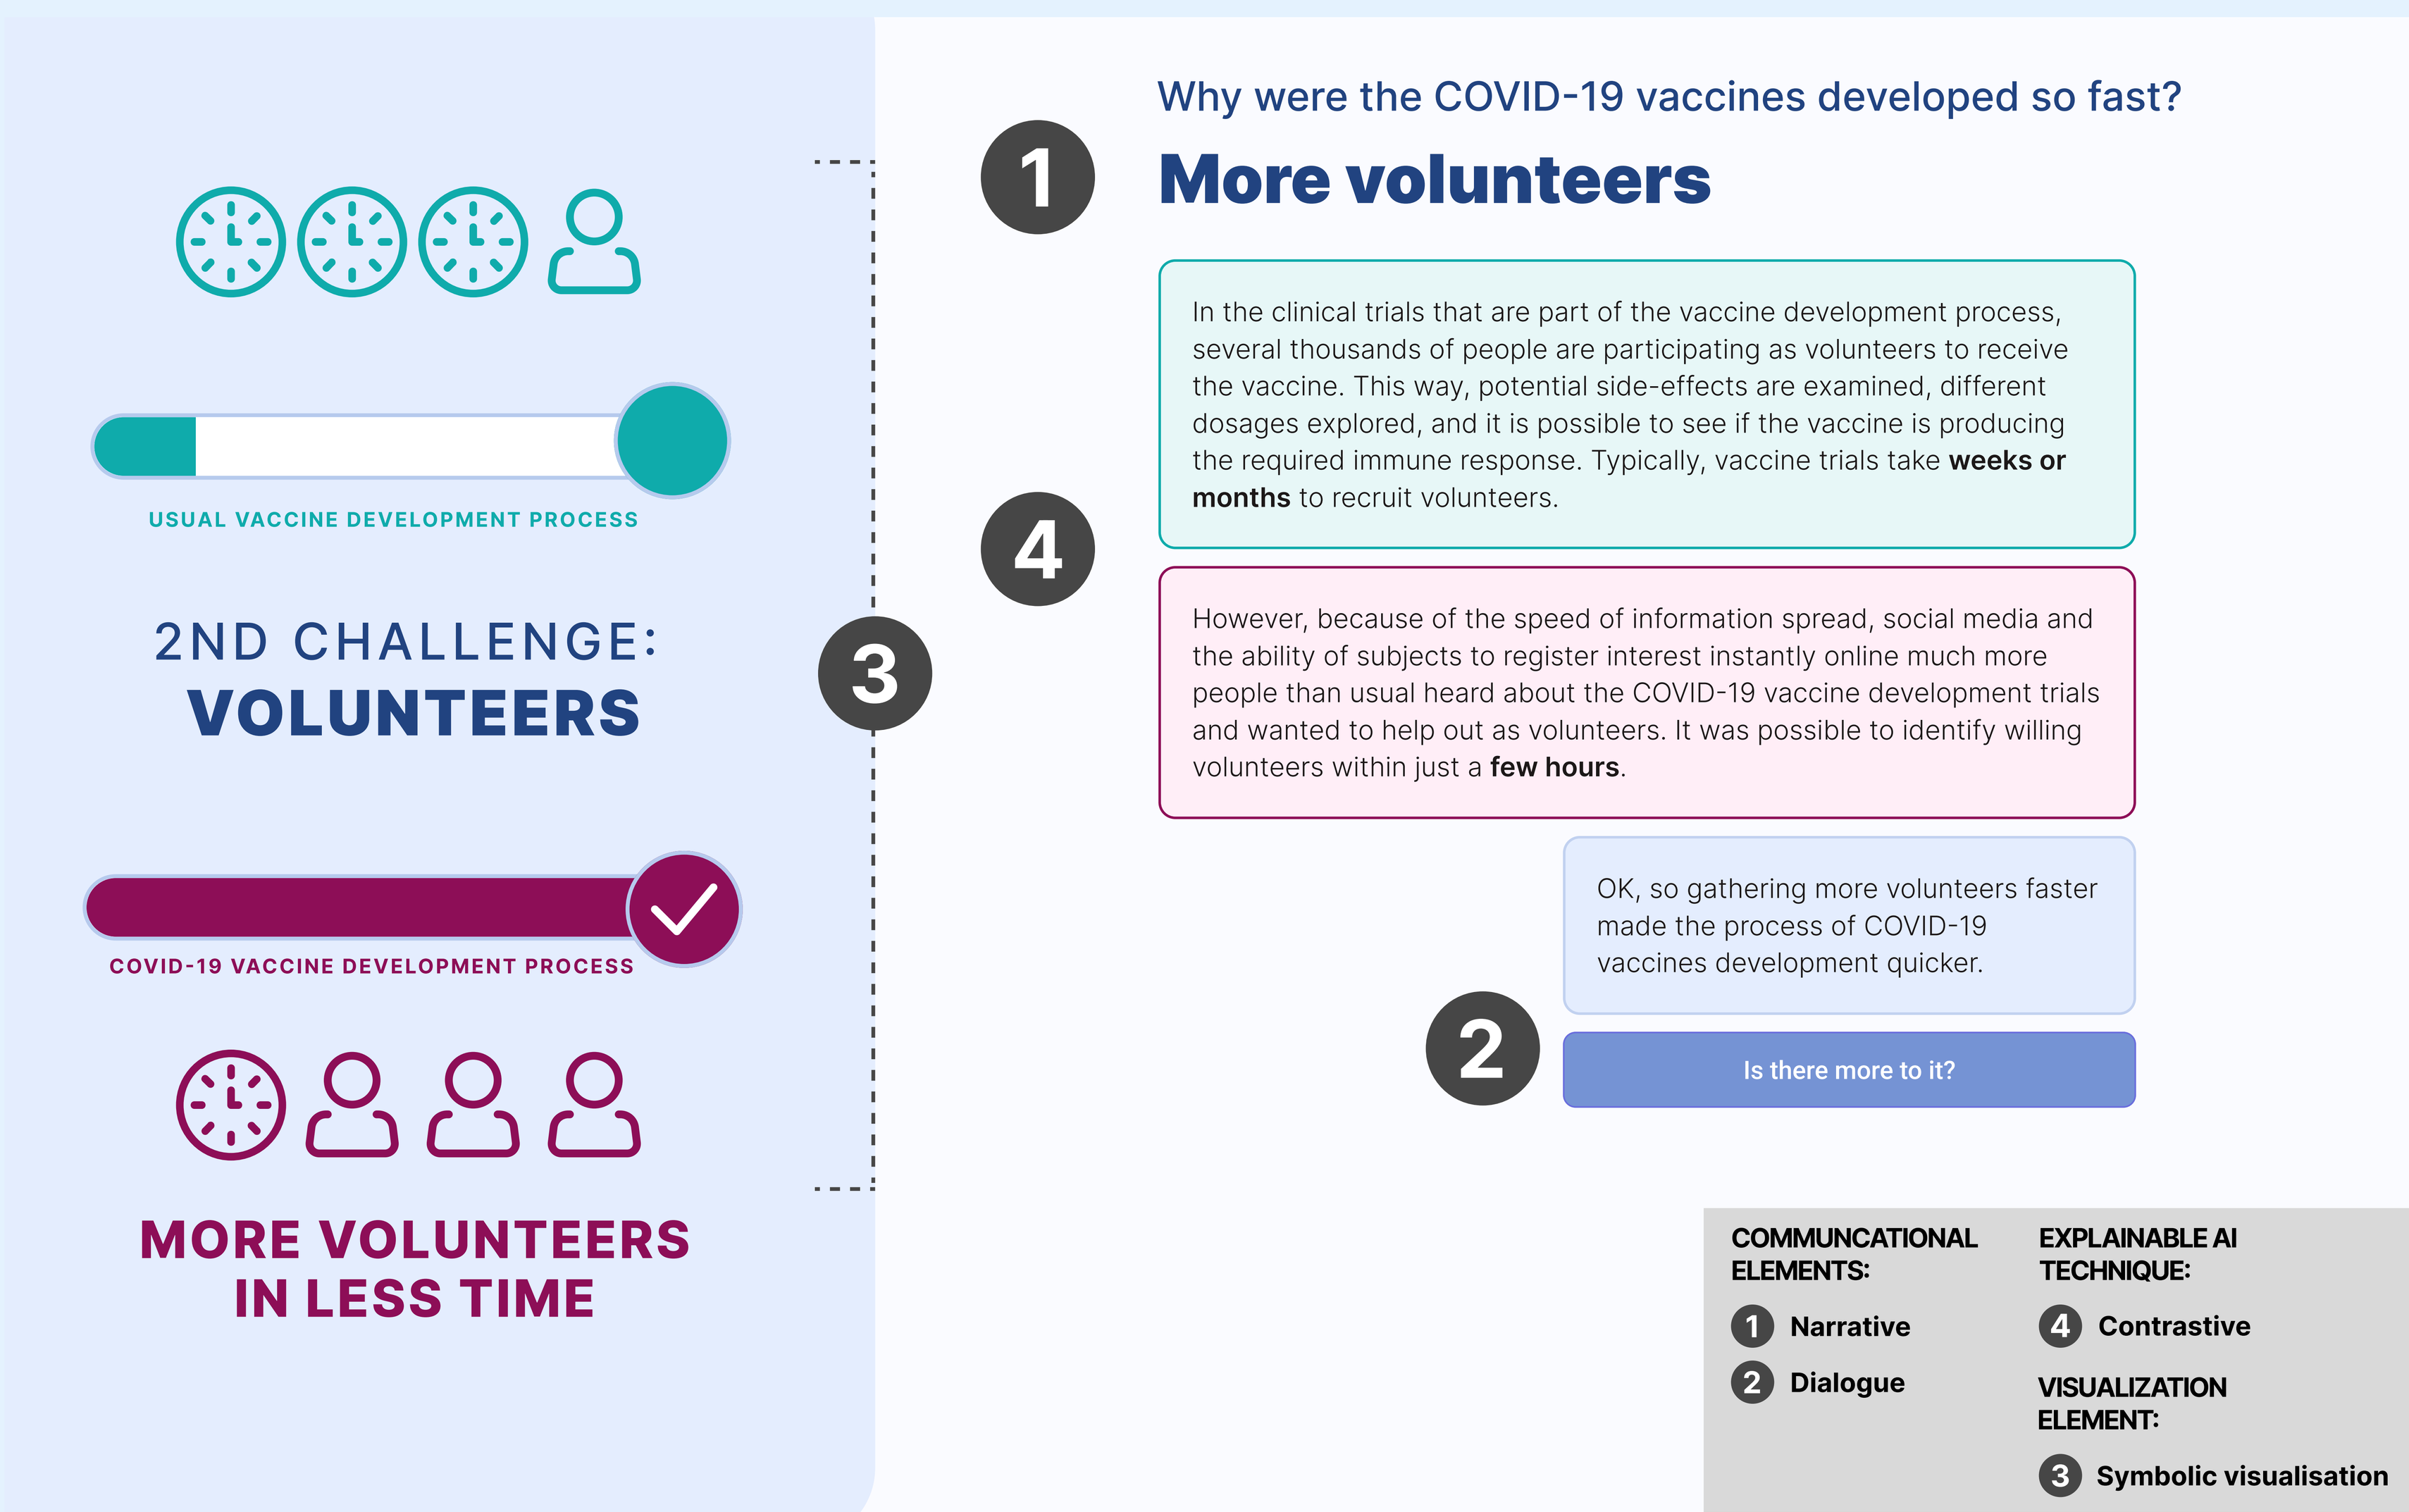

2ND CHALLENGE: VOLUNTEERS
USUAL VACCINE DEVELOPMENT PROCESS
COVID-19 VACCINE DEVELOPMENT PROCESS
MORE VOLUNTEERS IN LESS TIME
Why were the COVID-19 vaccines developed so fast?
More volunteers
In the clinical trials that are part of the vaccine development process, several thousands of people are participating as volunteers to receive the vaccine. This way, potential side-effects are examined, different dosages explored, and it is possible to see if the vaccine is producing the required immune response. Typically, vaccine trials take weeks or months to recruit volunteers.
However, because of the speed of information spread, social media and the ability of subjects to register interest instantly online much more people than usual heard about the COVID-19 vaccine development trials and wanted to help out as volunteers. It was possible to identify willing volunteers within just a few hours.
OK, so gathering more volunteers faster made the process of COVID-19 vaccines development quicker.
Is there more to it?
COMMUNCATIONAL ELEMENTS:
1 Narrative
2 Dialogue
EXPLAINABLE AI TECHNIQUE:
4 Contrastive
VISUALIZATION ELEMENT:
3 Symbolic visualisation

Figure 2. One of the four explanation steps with elements of the explanation mapped out.

Design Process

To address the need for better communicational tools for explaining complex COVID-19 vaccine-related information, we applied an **iterative human-centered process** similar to **Design Thinking**, consisting of 5 phases; **empathize**, **define**, **ideate**, **prototype** and **test**.

Phase I: Empathize

Goals:
- To gain an **emphatic understanding** of the problem by combining a **scientific literature** analysis with a **human-centered** analysis.

How:
- Scientific problem understanding was developed based on the **5C model of vaccine hesitancy factors** [1].
- For human-centered understanding **we turned to people directly**, asking them about their vaccine-related concerns, ways they informed themselves, and how they perceived materials covering these issues. We also analyzed a sample of **media articles**, **discussions in online forums**, and **interviews** available on YouTube.

Phase II: Define

Goal:
- Define **one core problem** in need of an explanation guided by the **users' framing** of the problem.

How:
- **Personas** were created based on findings from the Empathize phase, each representing one group and its most common COVID-19 vaccine concerns.
- Important insight: **one solution does not fit all**, because general explanations do not answer specific concerns people have.
- Core problem definition: **"How could COVID-19 vaccines be safe if they have been developed so quickly – unlike previous vaccines that took much longer?"**.

Phase III: Ideate

Goals:
- Ideate **possible solutions** on how to explain the defined problem.

How:
- The core of our conceptual solution design was **a contrastive** explanation technique from explainable AI - answering the question "**Why outcome P, rather than outcome Q has occurred?**" (read more about explainable AI techniques in [4]).
- Elements of **narrative** exposition and **parasocial** identification [5], **motivational interviewing** [3], and **metaphorical** and **symbolic** visualizations [2] were integrated to the conceptual solution.

Phase IV: Prototype

Goal:
- **Explore** how to **implement different elements** to an explanation.

How:
- Through an iterative process.
- We created an interactive web-based explanation based on the conceptual solution from the Ideation phase (see screenshots: Figure 1 & 2).
- The realization of the prototype is in a form of a **guided dialogue** with a **scientist character**, who explains **four major differences** between the COVID-19 vaccine development process, and the usual one (more funding, volunteers & data; less bureaucracy).

Phase V: Test

Goal:
- **Evaluate** the overall solution concept and **obtain feedback** for **improving** the prototype.

How:
- **Semi-structured think-aloud interviews** were conducted with 6 target users.
- All participants found the explanation prototype **easy to understand**, **well-structured**, and **written in a user-friendly language**.
- All interviewees stated more interactivity and **adding further visualizations** could provide an even better understanding.

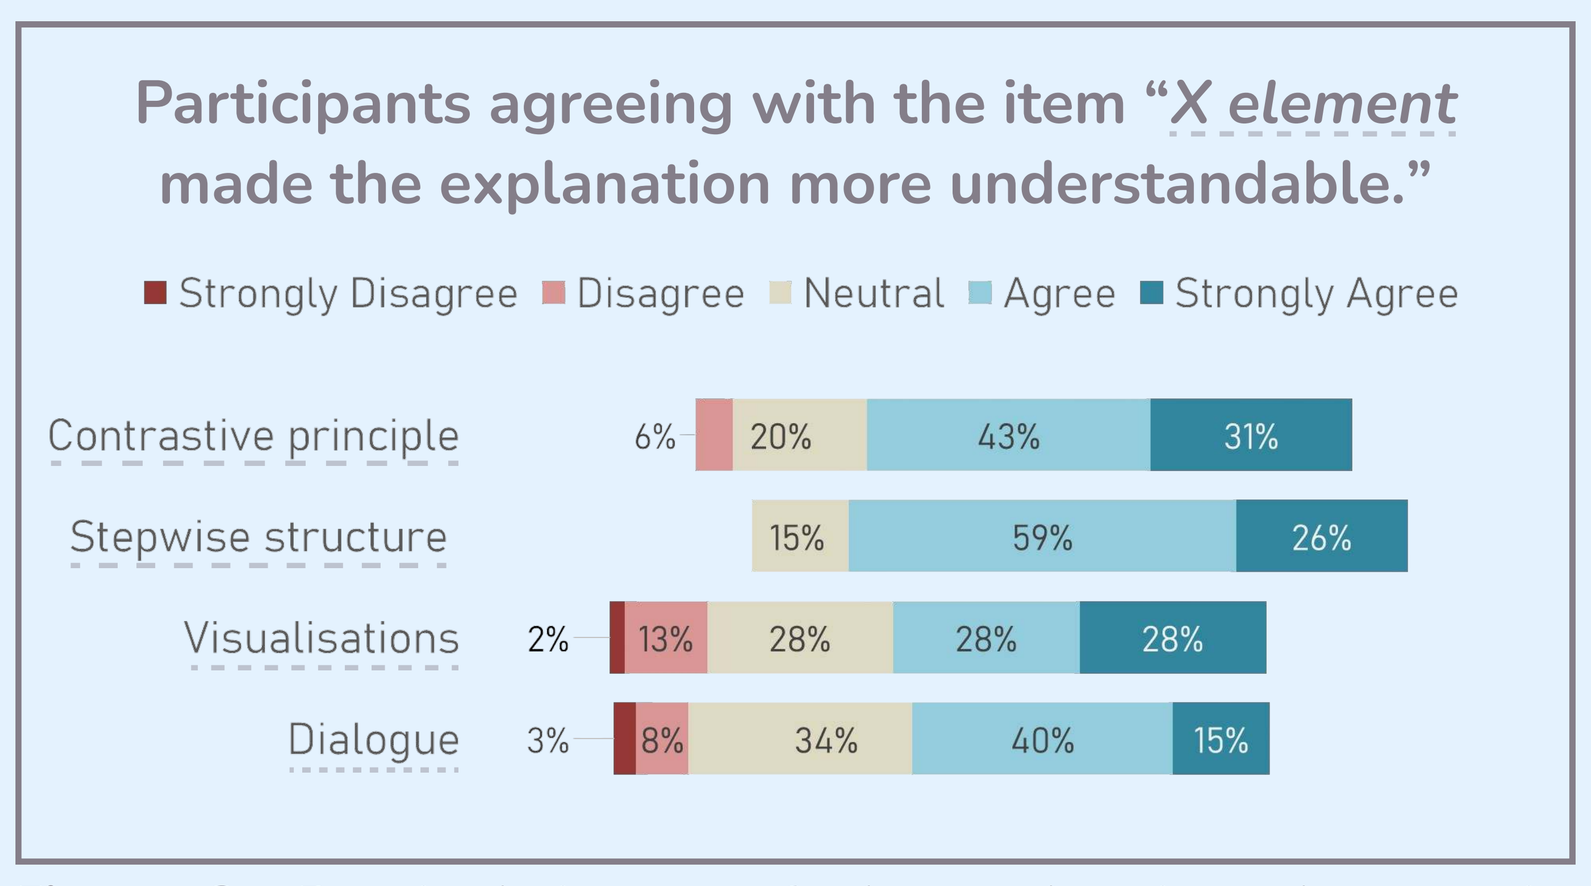

Participants agreeing with the item "X element made the explanation more understandable."
Strongly Disagree
Disagree
Neutral
Agree
Strongly Agree
Contrastive principle 6% 20% 43% 31%
Stepwise structure 15% 59% 26%
Visualisations 2% 13% 28% 28% 28%
Dialogue 3% 8% 34% 40% 15%

Figure 3. Perceived impact of the explanation elements its understandability. N = 88

Evaluation & Insights

The developed prototype was evaluated through an **online survey** during two interactive online workshops; one with bachelor students in health communication, the second one with high-school students. The survey was completed by 45 bachelor students (69% female, 29% male, age 19-28), and 43 high-school students (60% female, 21% male, 9% diverse, age 15-18).

82.5% agreed that the explanation is **understandable**

68% agreed that the concern was **well addressed**

48% agreed that the prototype could **increase the willingness to vaccinate**

55% would **share the prototype with a vaccine hesitant person**

Conclusions & Future Work

While being careful not to draw too strong conclusions from this explorative evaluation, the results indicate that **the prototype could have a positive impact on the openness of users towards considering the information presented in the explanation**.

Based on insights gathered during the development and evaluation of the presented prototype, **we have created one more explanation** of another common concern related to the COVID-19 crises, one that is present especially lately – **"Why is it still a good idea to wear a mask?"**. This prototype is **more interactive**, accompanied by **more visuals**, and has the property of being even **more tailored** and **adjustable** to different people's need for explanations. The evaluation of the said prototype is done and we are currently in the phase of interpreting the results, which look promising.

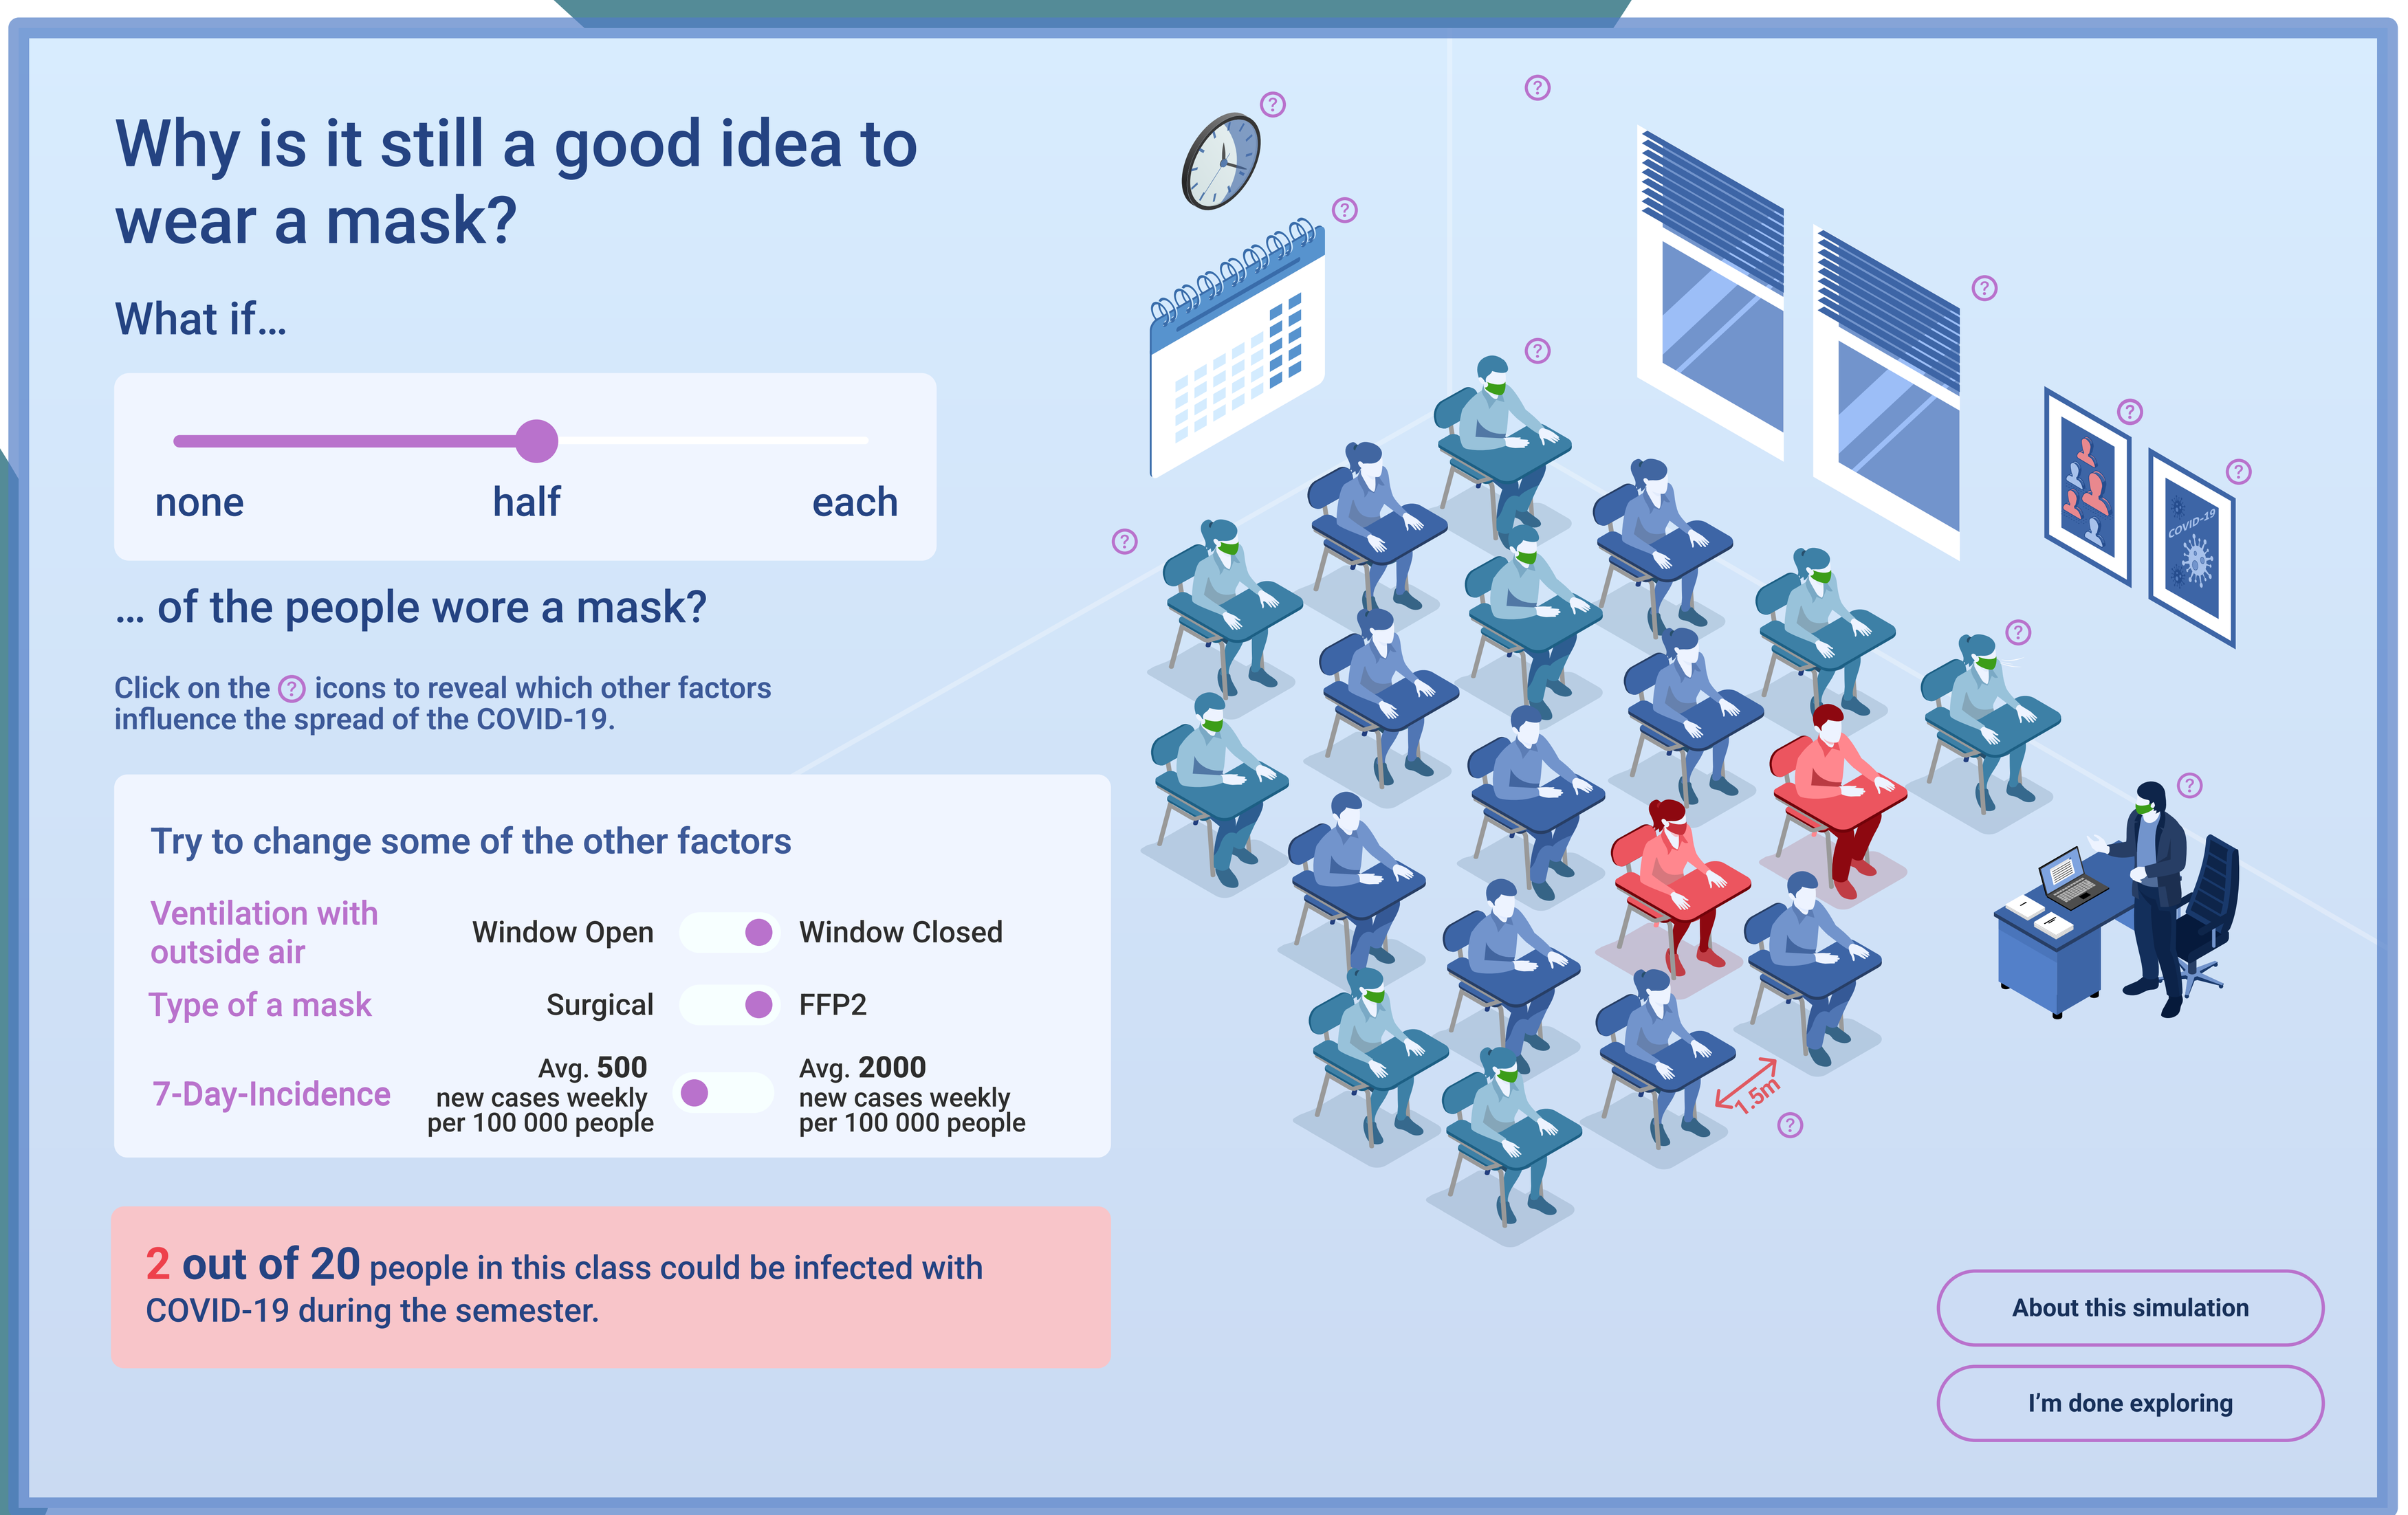

Why is it still a good idea to wear a mask?
What if...
none
half
each
... of the people wore a mask?
Click on the ? icons to reveal which other factors influence the spread of the COVID-19.
Try to change some of the other factors
Ventilation with outside air
Window Open
Window Closed
Type of a mask
Surgical
FFP2
7-Day-Incidence
Avg. 500 new cases weekly per 100 000 people
Avg. 2000 new cases weekly per 100 000 people
2 out of 20 people in this class could be infected with COVID-19 during the semester.
1.5m
About this simulation
I'm done exploring

References

[1] Betsch C, Schmid P, Heinemeier D, Korn L, Holtmann C, Böhm R (2018) Beyond confidence: Development of a measure assessing the 5C psychological antecedents of vaccination. PLoS ONE 13(12): e0208601. https://doi.org/10.1371/journal.pone.0208601

[2] Cominola, A., Giuliani, M., Castelletti, A., Fraternali, P., Herrera Gonzalez, S.L., Guardiola Herrero, J. C., Novak, J. & Emilio Rizzoli, A. (2021). Long-term water conservation is fostered by smart meter-based feedback and digital user engagement. npj Clean Water 4 (1), 1-10. https://doi.org/10.1038/s41545-021-00119-0

[3] Hettema, J., Steele, J., & Miller, W. R. (2005). Motivational interviewing. Annual Review of Clinical Psychology, 1, 91-111.

[4] Miller, T. (2018). Explanation in artificial intelligence: Insights from the social sciences. Artificial Intelligence, 267, 1-38.

[5] Petty, R. E. and Cacioppo, J. T. (1986). 'The Elaboration Likelihood Model of Persuasion'. In: Communication and Persuasion. New York, U.S.A.: Springer, pp. 1–24.

Stay updated
Scan the QR code to open the project web-page.

Read more
Scan the QR code to access the full-text article.

Explore
Scan the QR code to try out the new prototype.

EIPCM European Institute for Participatory Media
HOST Hochschule Stralsund University of Applied Sciences
Radboud University

The work presented on this poster has been funded by the Volkswagen Stiftung (grant nr: 97260-1). We also thank Prof. Dr. Enny Das and Prof. Dr. Martha Larson from Radbound University for their feedback and support of the project, as well as Boryana Krasimirova for her work on the visual and interaction design of the prototype.